Cancer Immunotherapy Biomarker Profiling Assay

CELLECTA
www.cellecta.com

Alex Chenchik[1], Mikhail Makhanov[1], Leonid Iakoubov[1], and Costa Frangou[1].
[1]Cellecta, Inc., Mountain View, CA USA

Introduction

Background:

- Link between the immune infiltrate and several human carcinoma types and prognosis and/or response to therapy
- Increasing evidence suggests that the number, type, and location of tumor-infiltrating lymphocytes in primary tumors harbor prognostic value, and this has led to the development of a "tumor immunoscore / immune" index

Aims:

- Custom and pre-made CancerNet expression assay services for analysis of cellular composition in tumor and blood samples
- Molecular profiling of key immune-related genes, including drug targets, known biomarkers, and immune mechanisms
- Unbiased discovery of most informative biomarkers that can be analyzed by conventional IHC/FACS assays

Applications:

- Detect cellular composition of immune / stromal / cancer cells in tumor microenvironment
- Identify immunity status and immunoediting mechanisms
- Discover novel biomarkers for immunotherapy
- Profiling of immunotherapy targets and all FDA-approved drug targets

CancerNet Assay Gene Composition

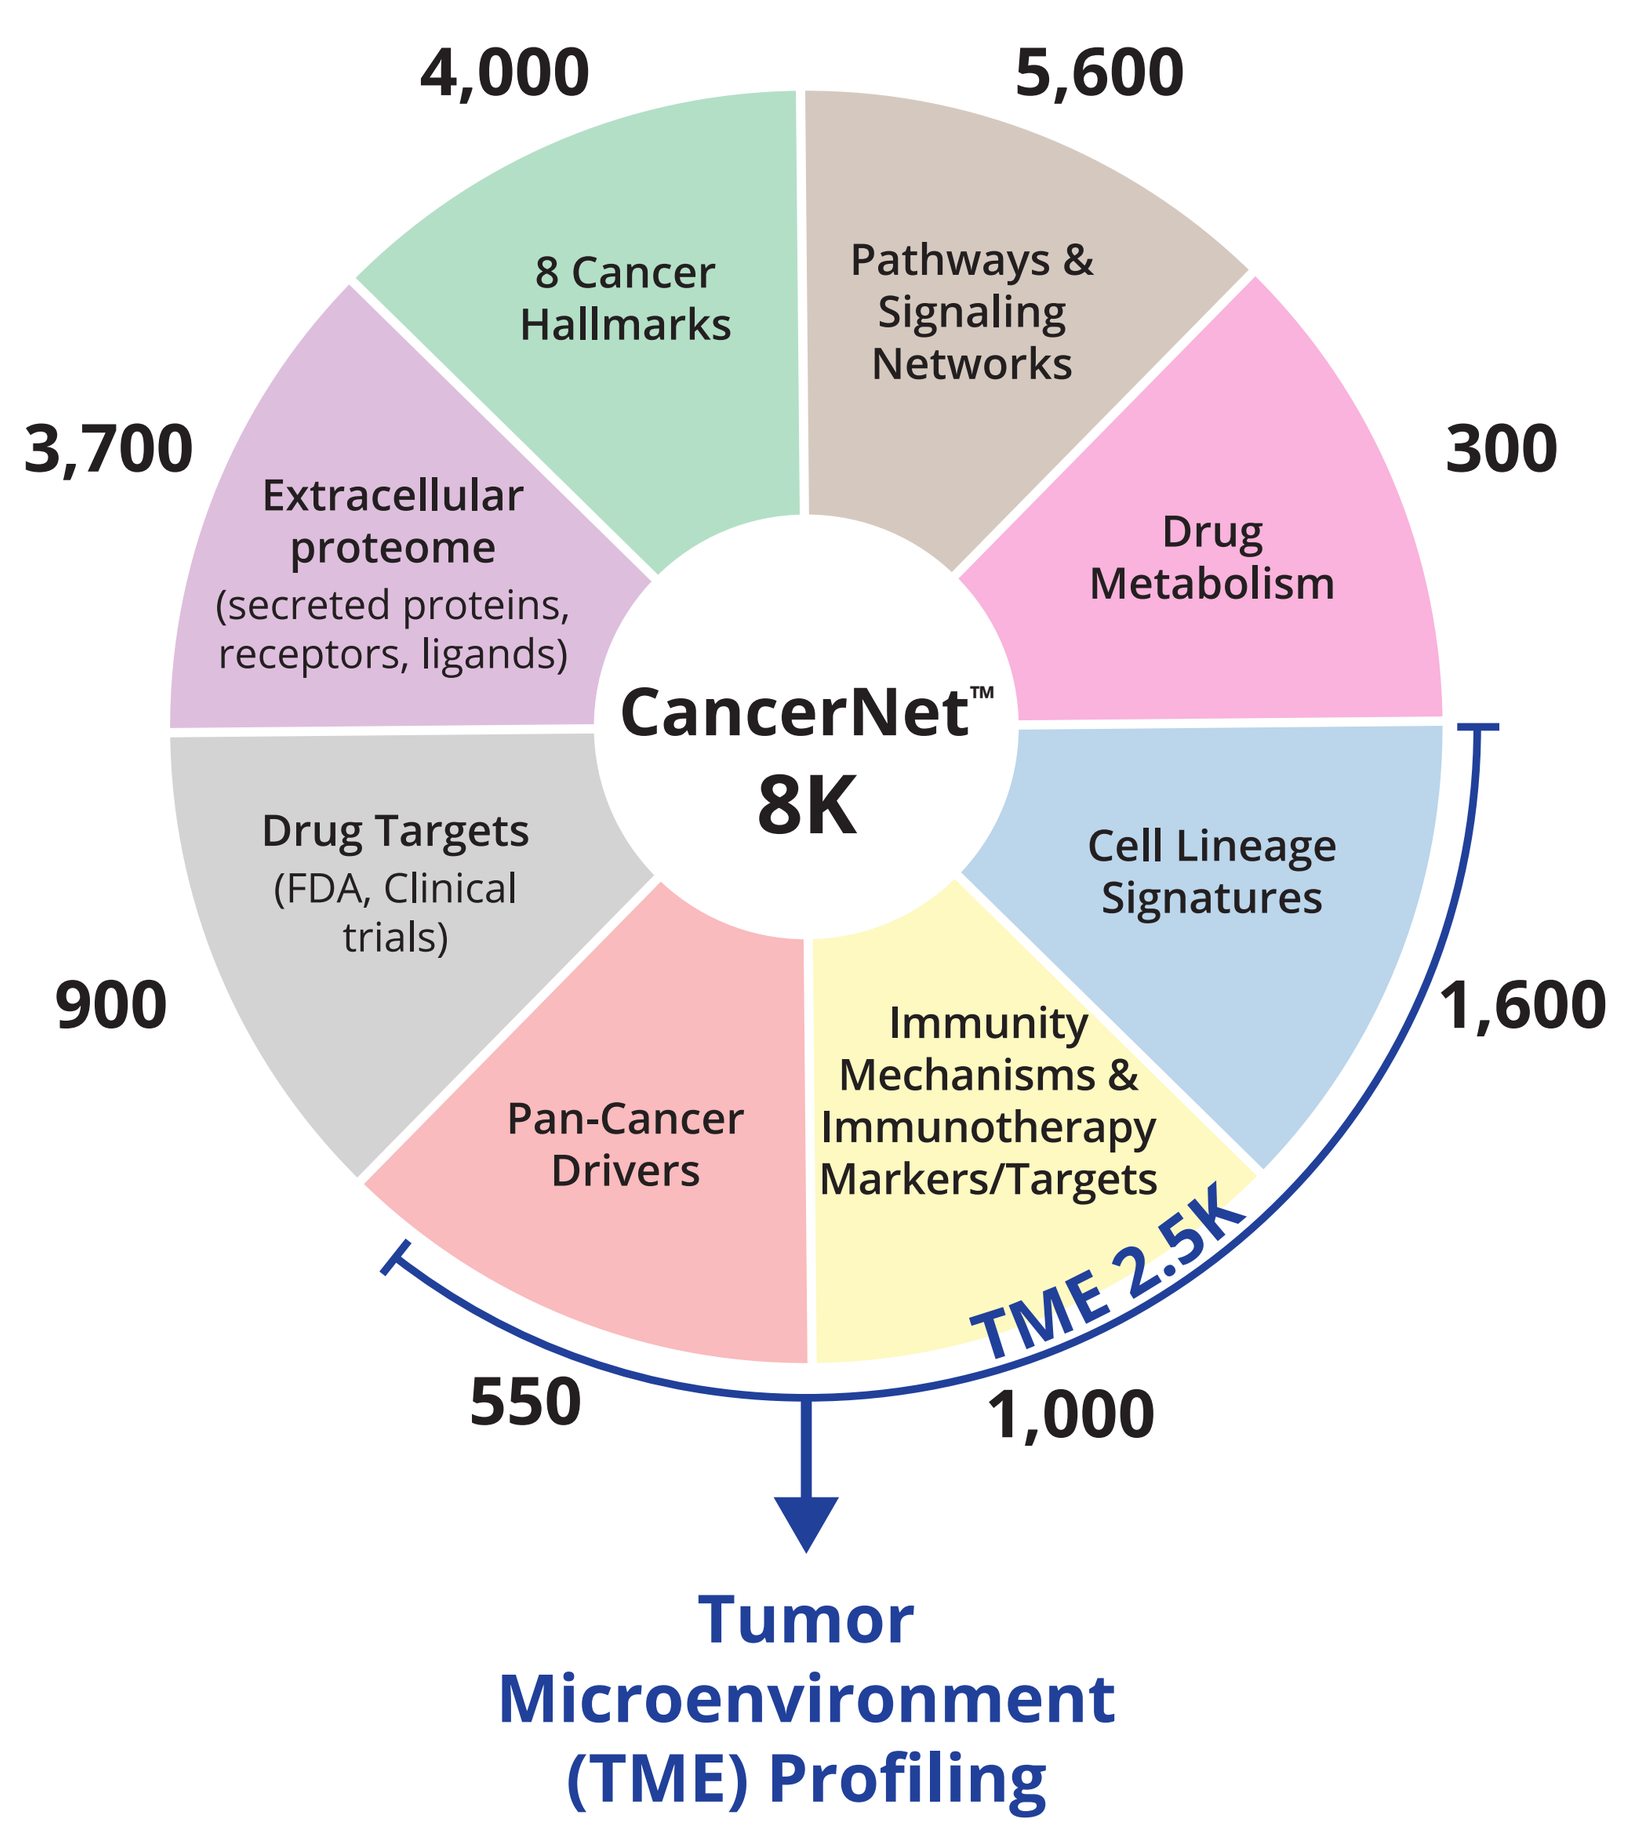
4,000
5,600
3,700
300
900
1,600
550
1,000
8 Cancer Hallmarks
Pathways & Signaling Networks
Drug Metabolism
Extracellular proteome (secreted proteins, receptors, ligands)
Drug Targets (FDA, Clinical trials)
Cell Lineage Signatures
Pan-Cancer Drivers
Immunity Mechanisms & Immunotherapy Markers/Targets
CancerNet™ 8K
TME 2.5K
Tumor Microenvironment (TME) Profiling

CancerNet Assay Workflow

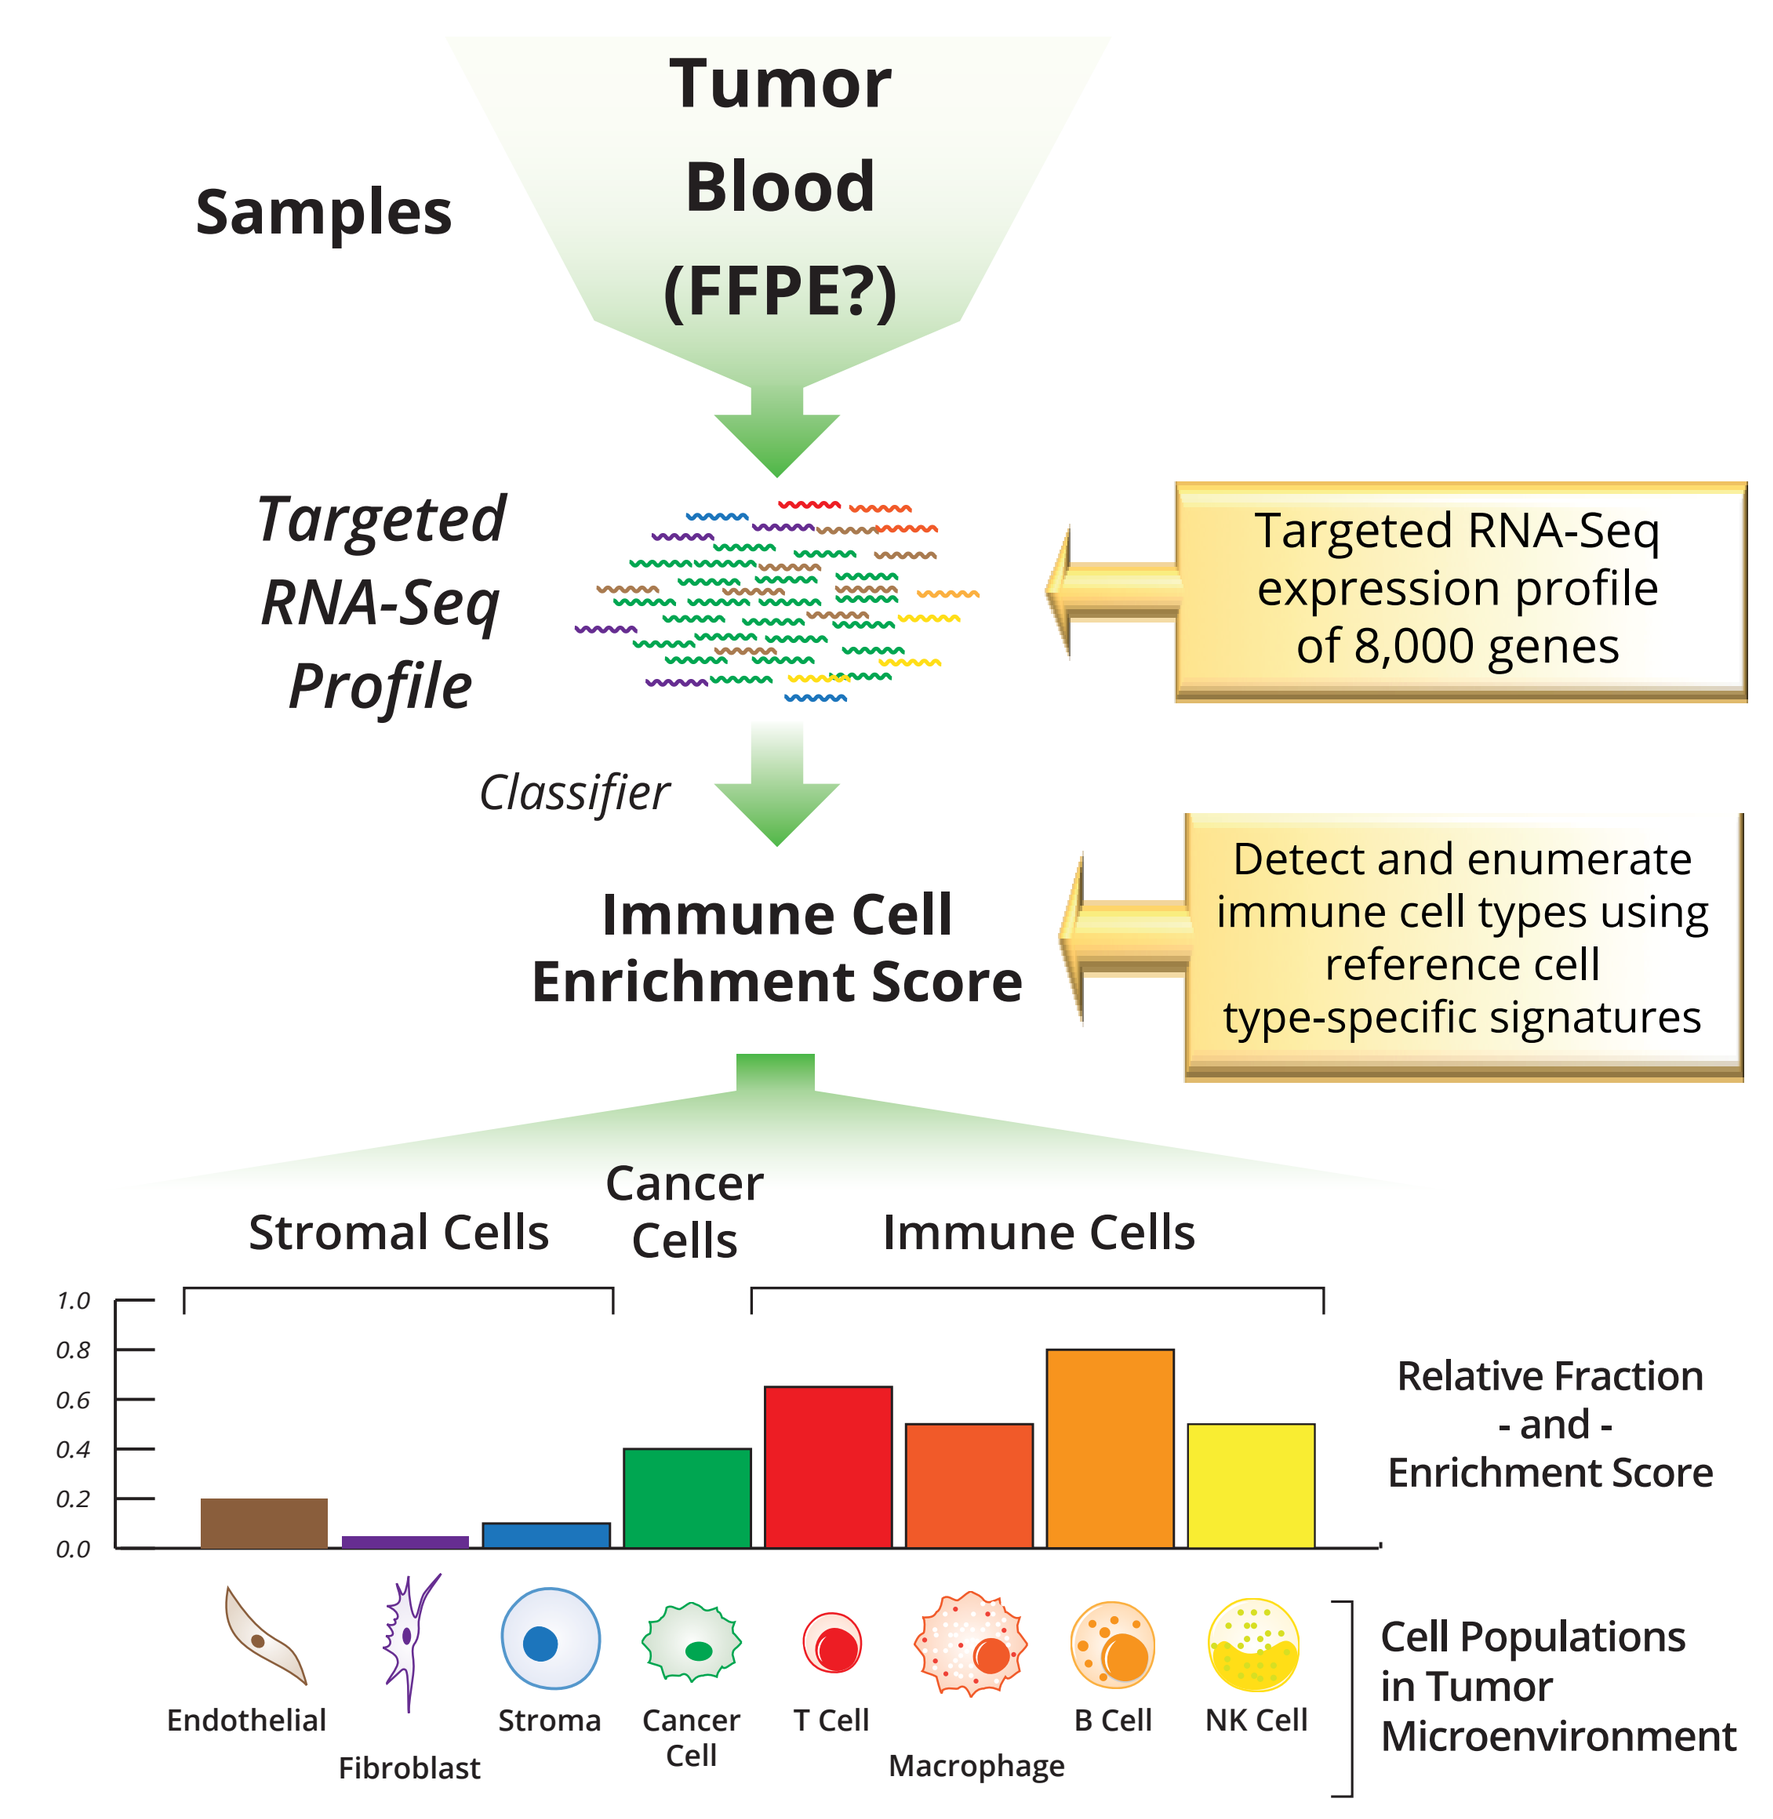
Samples
Tumor
Blood
(FFPE?)
Targeted RNA-Seq Profile
Targeted RNA-Seq expression profile of 8,000 genes
Classifier
Immune Cell Enrichment Score
Detect and enumerate immune cell types using reference cell type-specific signatures
Stromal Cells
Cancer Cells
Immune Cells
Relative Fraction - and - Enrichment Score
Endothelial
Fibroblast
Stroma
Cancer Cell
T Cell
Macrophage
B Cell
NK Cell
Cell Populations in Tumor Microenvironment

Summary of the CancerNet 8K pipeline and application to infiltrating tumor immune cell deconvolution. CancerNet uses a novel, multiplex, quantitative, targeted RNA-Seq assay and classifier algorithm to measure the absolute digital expression for ~2,500 genes and accurately resolve relative fractions of diverse immune cells types from complex tissues.

Development of Classifier Model for Different Immune Cell Types in Tumor Microenvironment

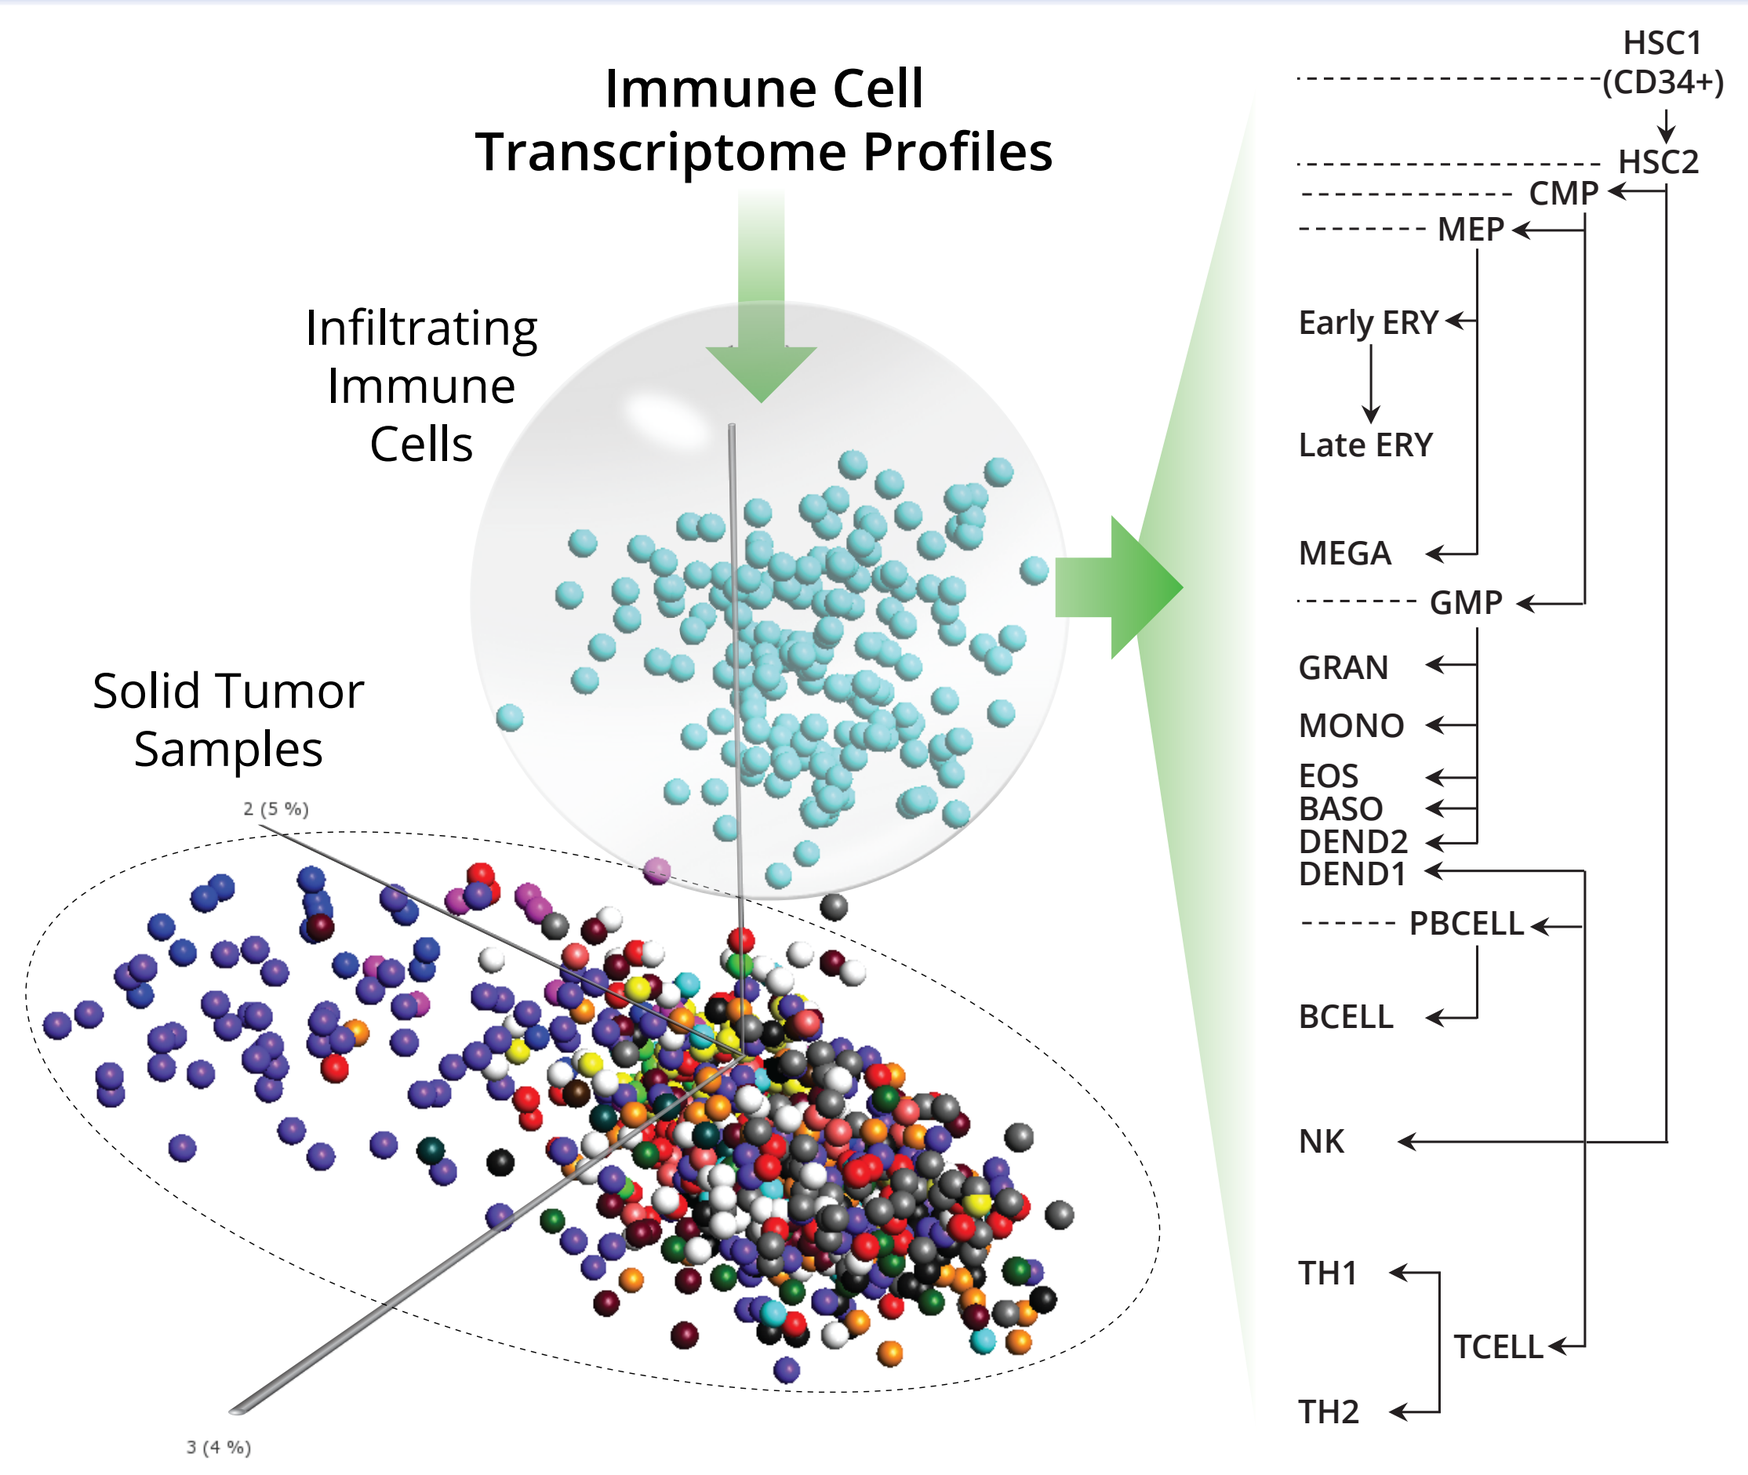
Immune Cell Transcriptome Profiles
Infiltrating Immune Cells
Solid Tumor Samples
HSC1 (CD34+)
HSC2
CMP
MEP
Early ERY
Late ERY
MEGA
GMP
GRAN
MONO
EOS
BASO
DEND2
DEND1
PBCELL
BCELL
NK
TH1
TCELL
TH2

Cluster Analysis with CancerNet 8K: Model reference gene expression data by principal component analysis using ImmuneNet differentially expressed (DE) genes successfully distinguishes hematopoietic cell phenotypes from bulk tissues and tumor cells. Diagram to right describes major cell types detected by the assay.

Development of Functionally Validated RT-PCR Primers

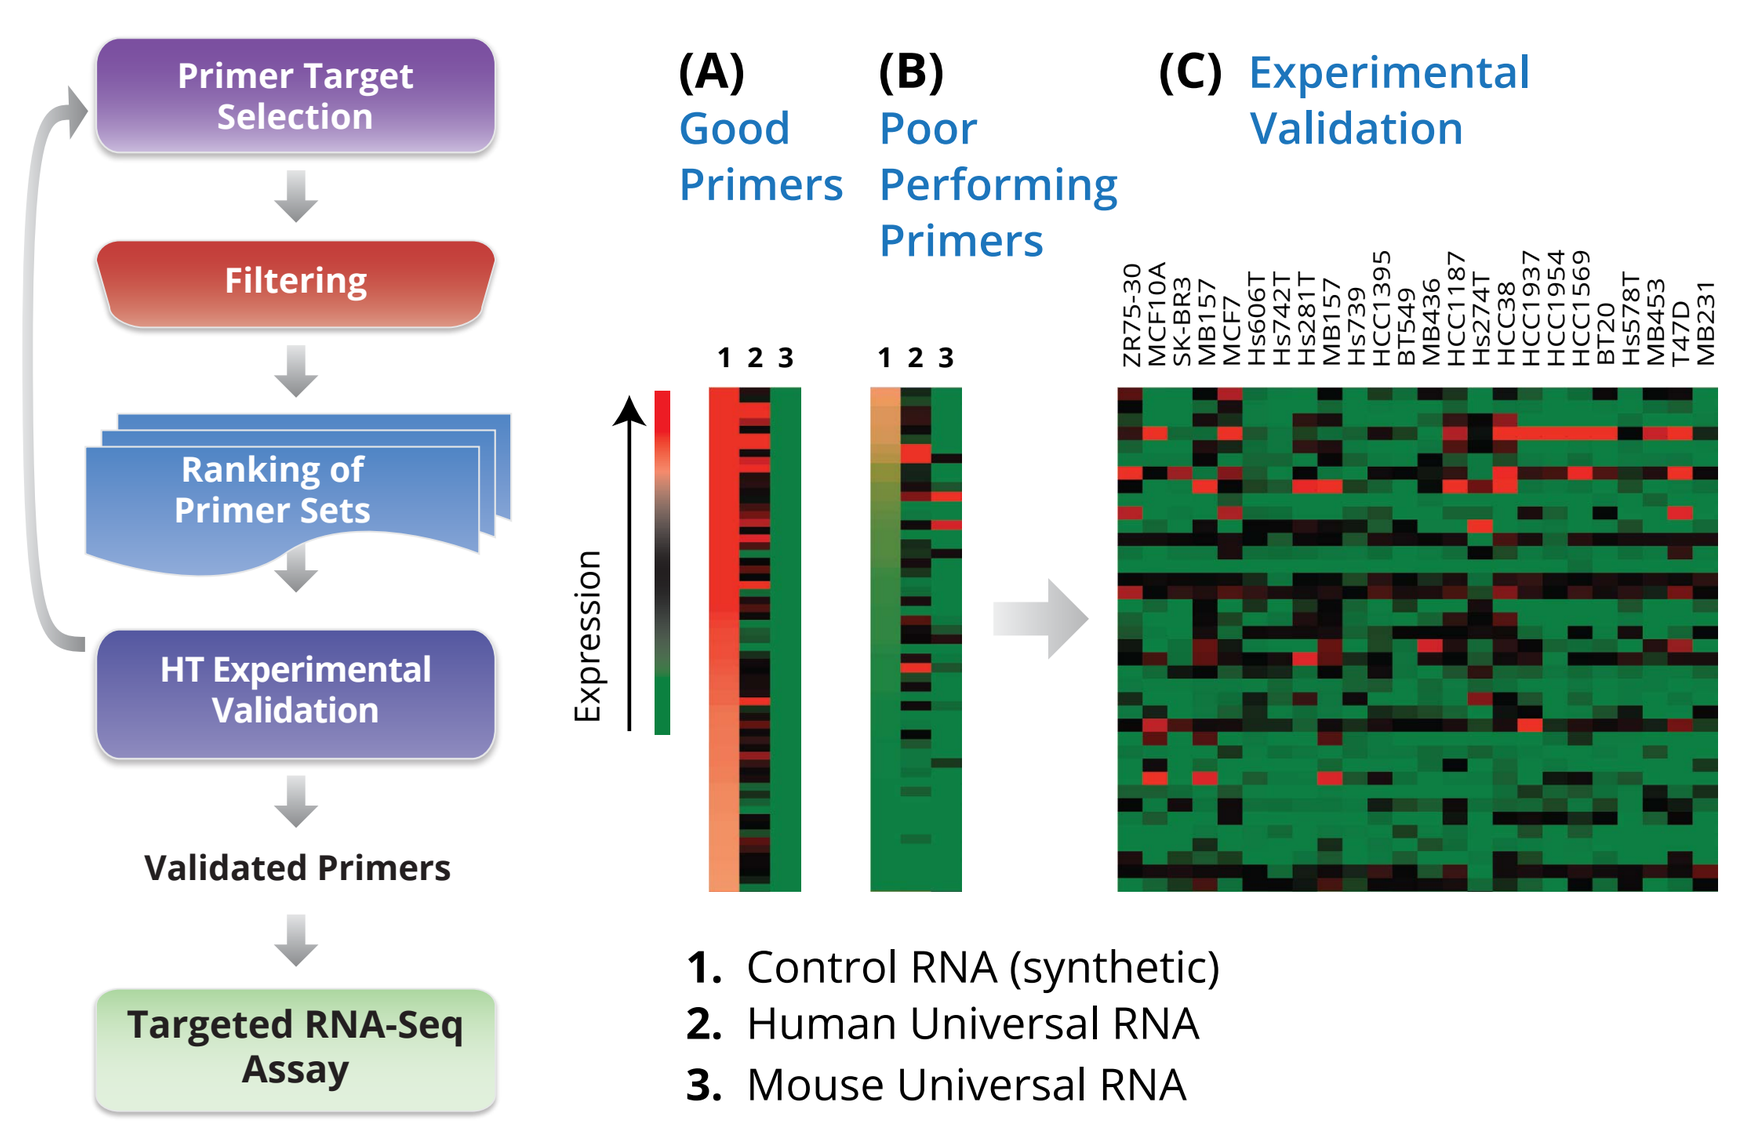
Primer Target Selection
Filtering
Ranking of Primer Sets
HT Experimental Validation
Validated Primers
Targeted RNA-Seq Assay
(A) Good Primers
(B) Poor Performing Primers
(C) Experimental Validation
Expression
1. Control RNA (synthetic)
2. Human Universal RNA
3. Mouse Universal RNA

Testing Primer Efficiency. Primers designed for the select panel of genes are functionally validated to be compatible with our high-throughput, multiplex protocol. Synthetic RNA control (Lane 1) and Human Universal RNA control (Lane 2) help demonstrate that the primers work efficiently in (A) and poorly as in (B). Mouse Universal RNA control (Lane 3) denotes cross-reactivity. Validated Primers with greater efficiency are selected and are subsequently used in the Targeted RNA-Seq assay. 24 cancer model cell lines were profiled against a select panel of genes (125). Red denotes upregulated genes and green denotes low expression in the heat-map presented (C).

Next-Generation Targeted RNA-Seq

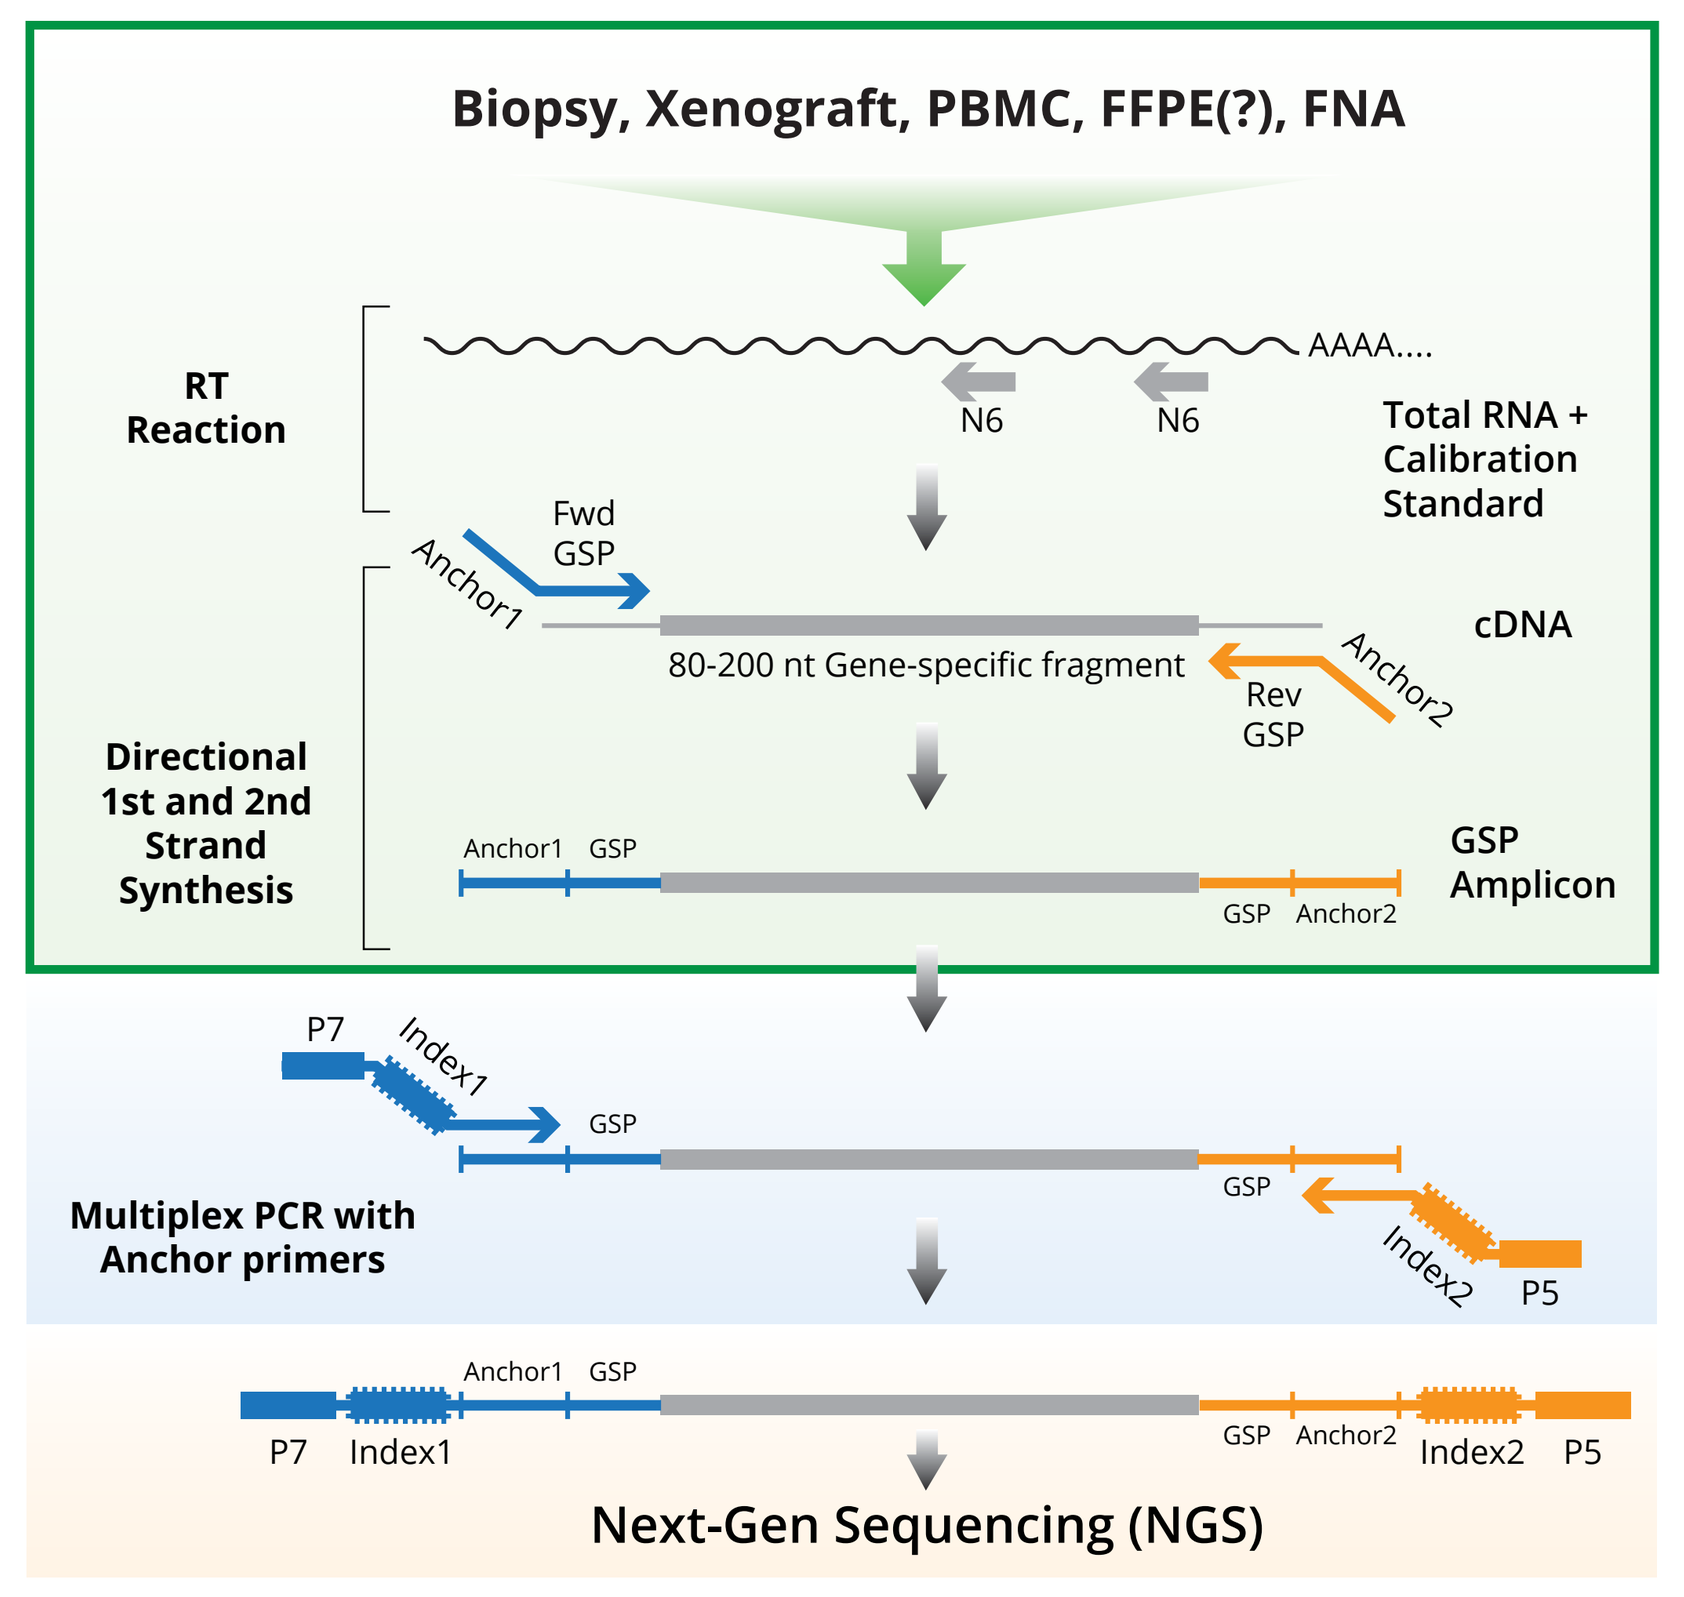
Biopsy, Xenograft, PBMC, FFPE(?), FNA
RT Reaction
Total RNA + Calibration Standard
cDNA
80-200 nt Gene-specific fragment
Directional 1st and 2nd Strand Synthesis
GSP Amplicon
Multiplex PCR with Anchor primers
Next-Gen Sequencing (NGS)

- Profiling of up to 10,000 genes in a single tube assay
- Requires 10-100 ng of total RNA from biopsy, blood, FFPE(?), or FNA samples
- Built-in calibration standards for QC and normalization of expression data

Our novel workflow leverages the power of quantitative PCR (upstream) and Next-Gen Sequencing (downstream read-out). Experimentally validated gene-specific primers are used to amplify selected target regions. Synthetic calibration controls act as an effective QC metric.

Validation of CancerNet Assay with TNBC Samples

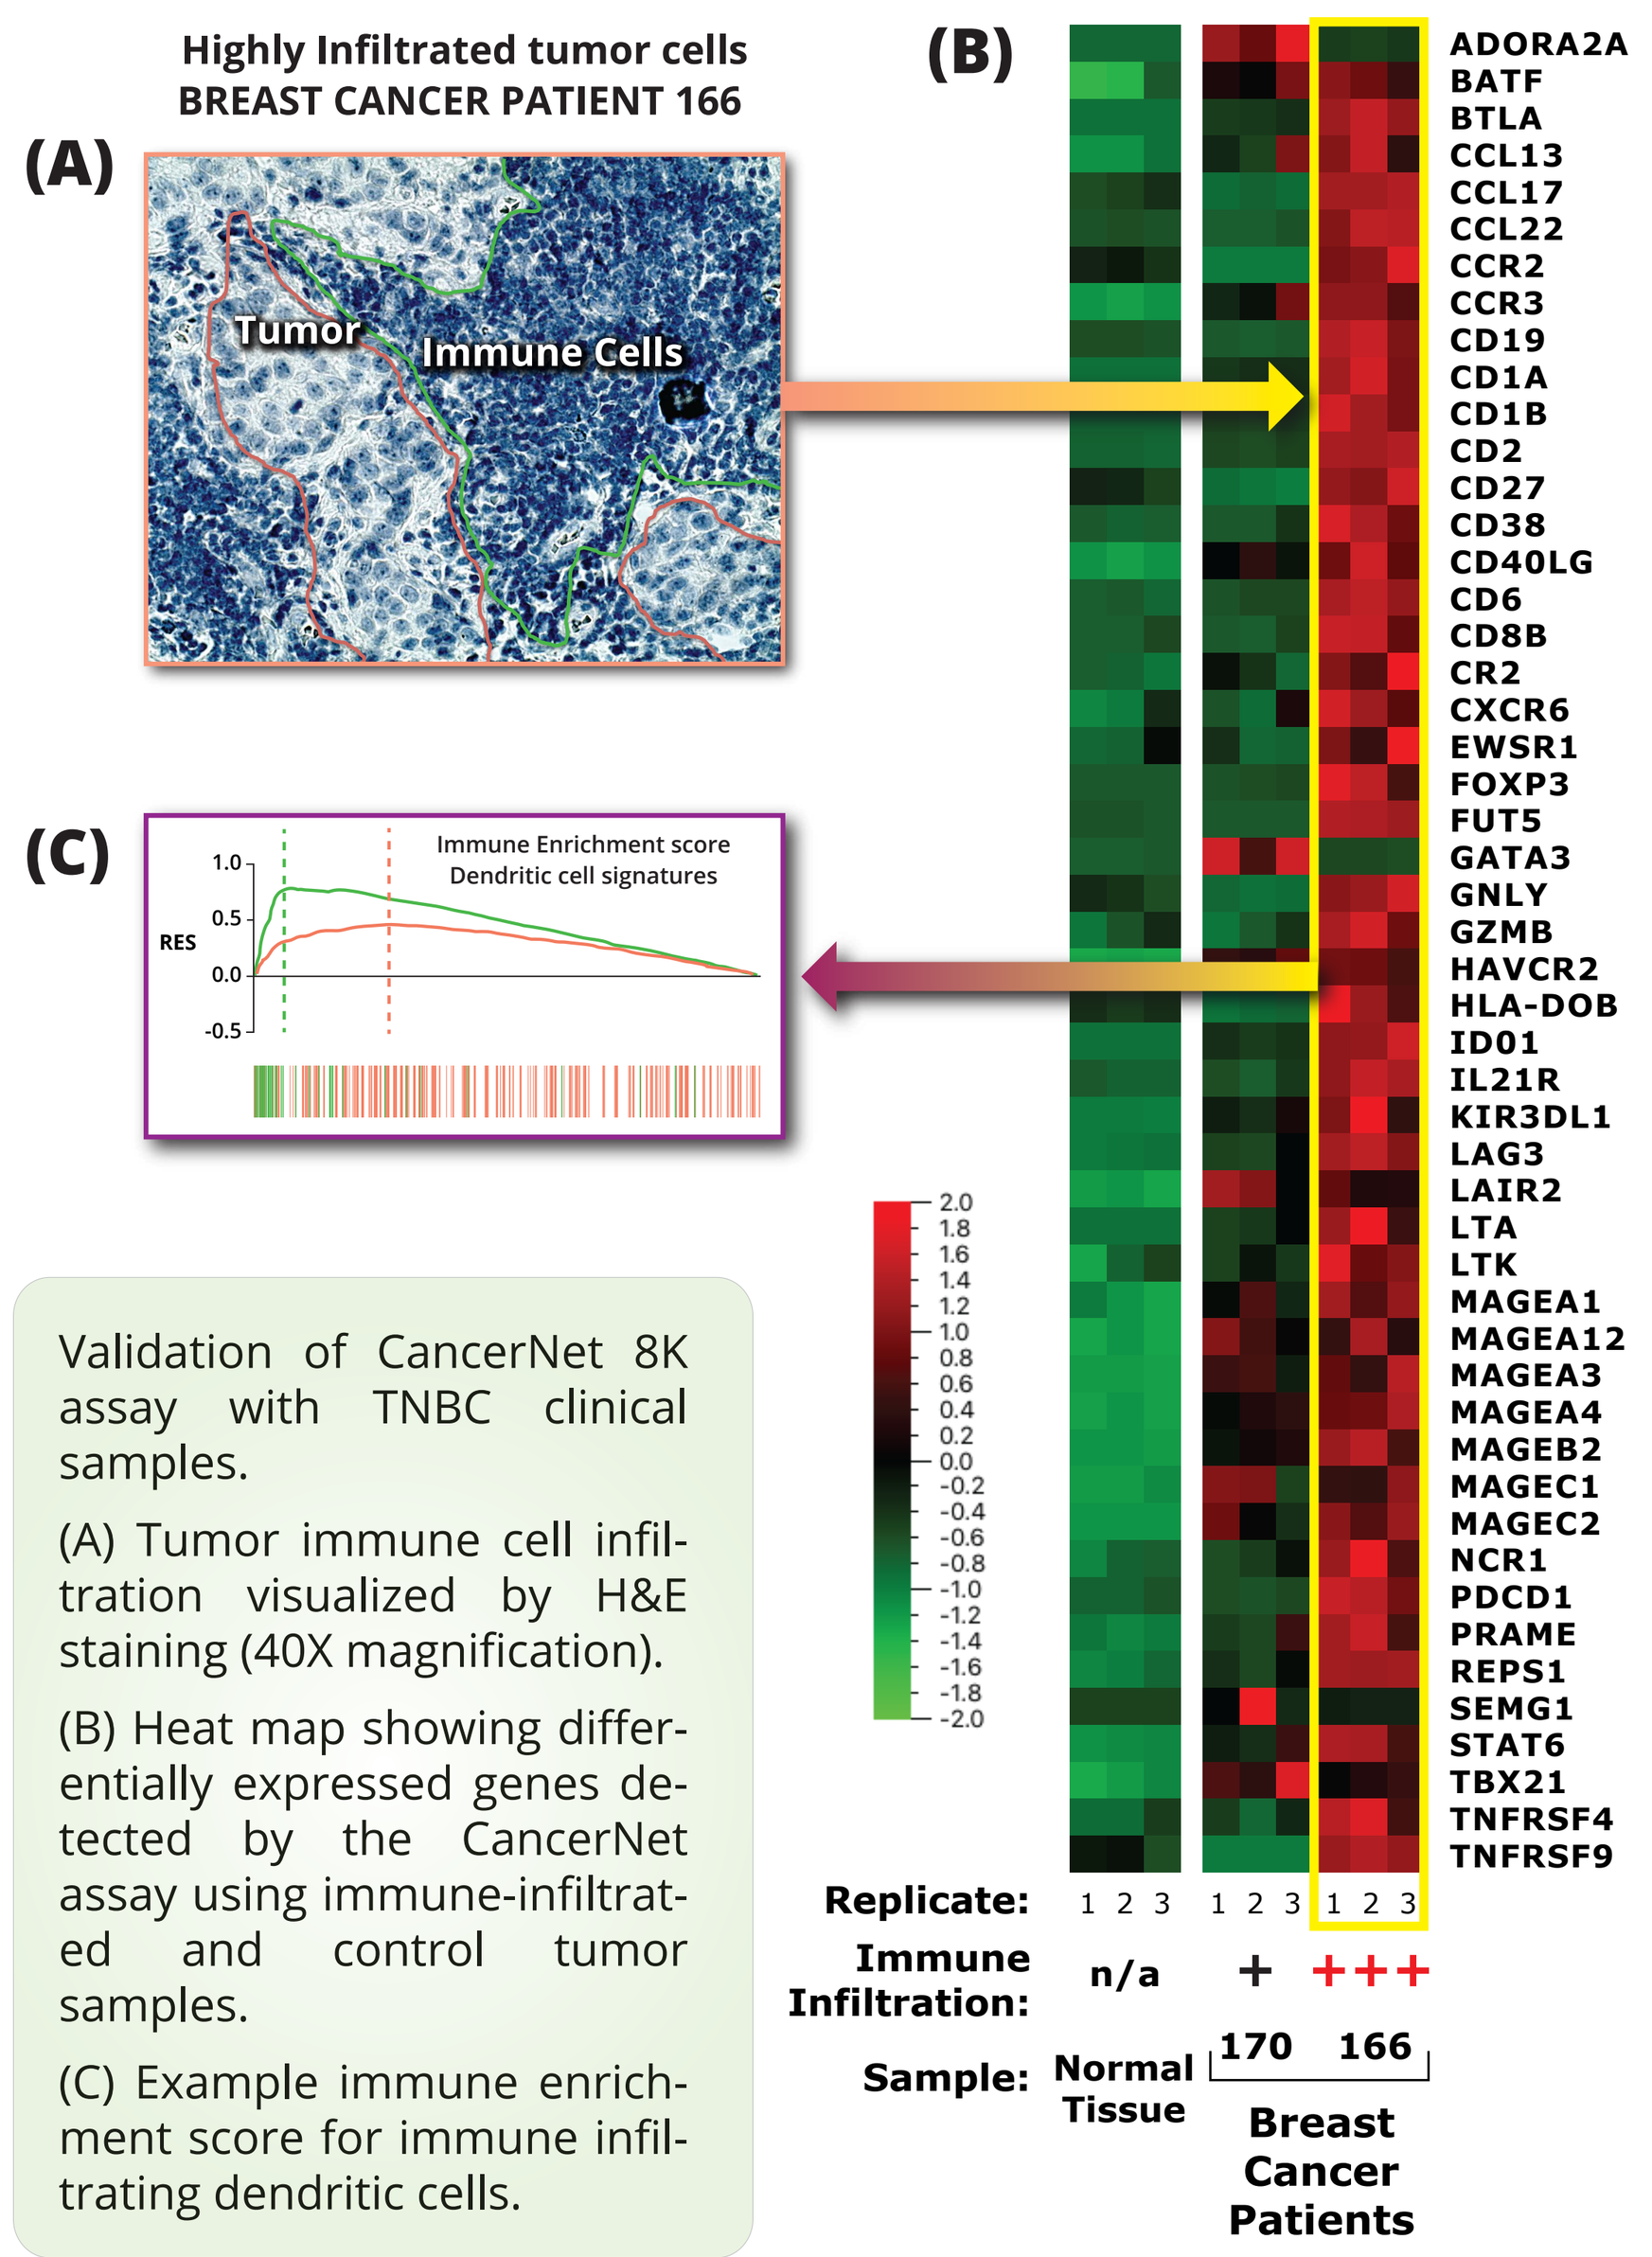
Highly Infiltrated tumor cells
BREAST CANCER PATIENT 166
(A)
Tumor
Immune Cells
(B)
(C)
Immune Enrichment score
Dendritic cell signatures
ADORA2A
BATF
BTLA
CCL13
CCL17
CCL22
CCR2
CCR3
CD19
CD1A
CD1B
CD2
CD27
CD38
CD40LG
CD6
CD8B
CR2
CXCR6
EWSR1
FOXP3
FUT5
GATA3
GNLY
GZMB
HAVCR2
HLA-DOB
IDO1
IL21R
KIR3DL1
LAG3
LAIR2
LTA
LTK
MAGEA1
MAGEA12
MAGEA3
MAGEA4
MAGEB2
MAGEC1
MAGEC2
NCR1
PDCD1
PRAME
REPS1
SEMG1
STAT6
TBX21
TNFRSF4
TNFRSF9
Replicate:
Immune Infiltration: n/a + +++
Sample: Normal Tissue 170 166 Breast Cancer Patients

Validation of CancerNet 8K assay with TNBC clinical samples.

(A) Tumor immune cell infiltration visualized by H&E staining (40X magnification).

(B) Heat map showing differentially expressed genes detected by the CancerNet assay using immune-infiltrated and control tumor samples.

(C) Example immune enrichment score for immune infiltrating dendritic cells.

Separation of Cell Type Specific Signatures

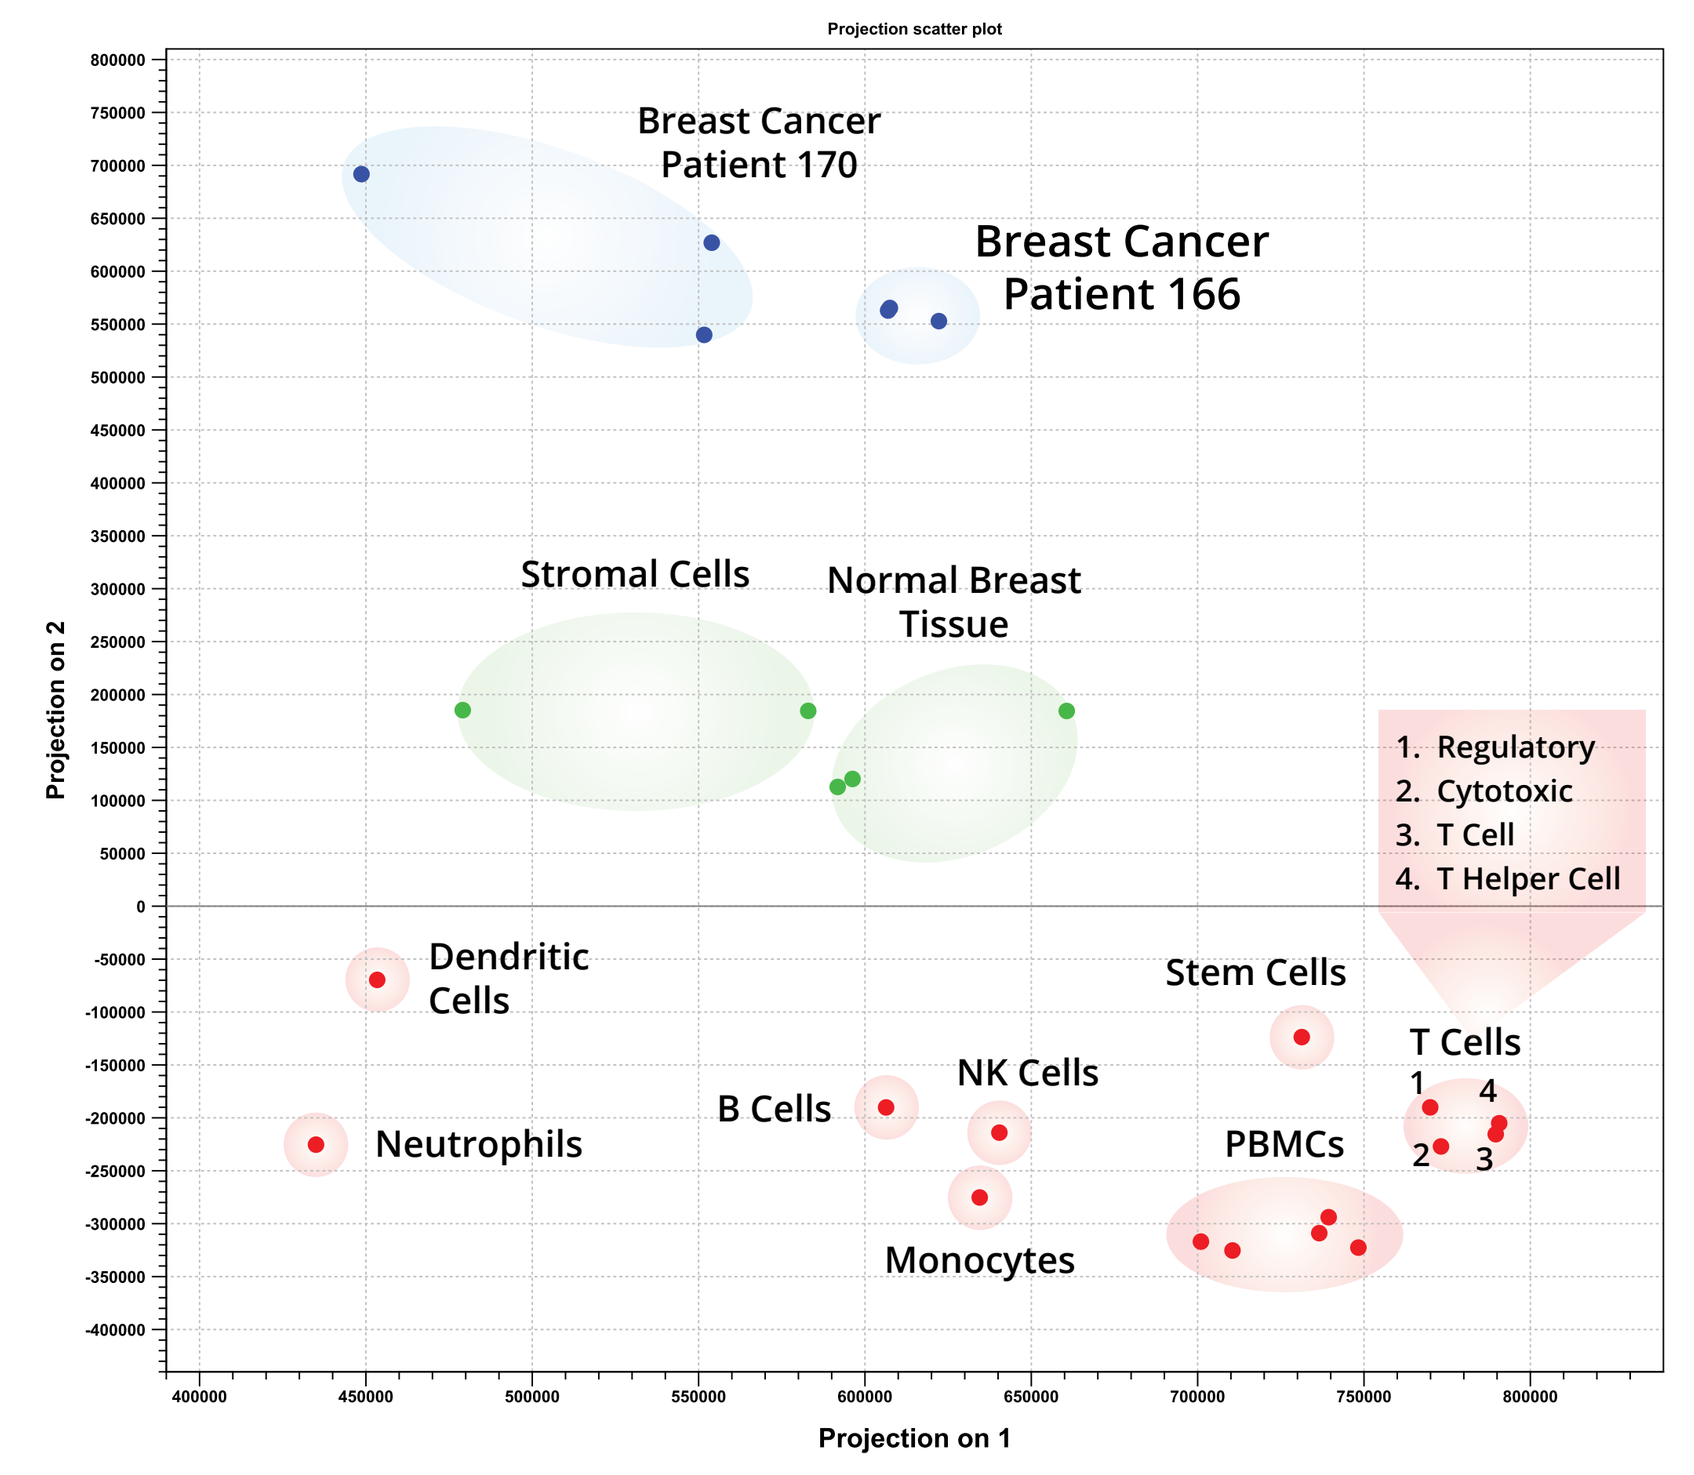
Projection scatter plot
Breast Cancer Patient 170
Breast Cancer Patient 166
Stromal Cells
Normal Breast Tissue
1. Regulatory
2. Cytotoxic
3. T Cell
4. T Helper Cell
Dendritic Cells
Stem Cells
T Cells
Neutrophils
B Cells
NK Cells
PBMCs
Monocytes
Projection on 1
Projection on 2

Principal Component Analysis of CancerNet cell type specific signatures for breast cancer, normal tissues, and reference immune and stromal cell types.

Reproducibility and Sensitivity of Assay

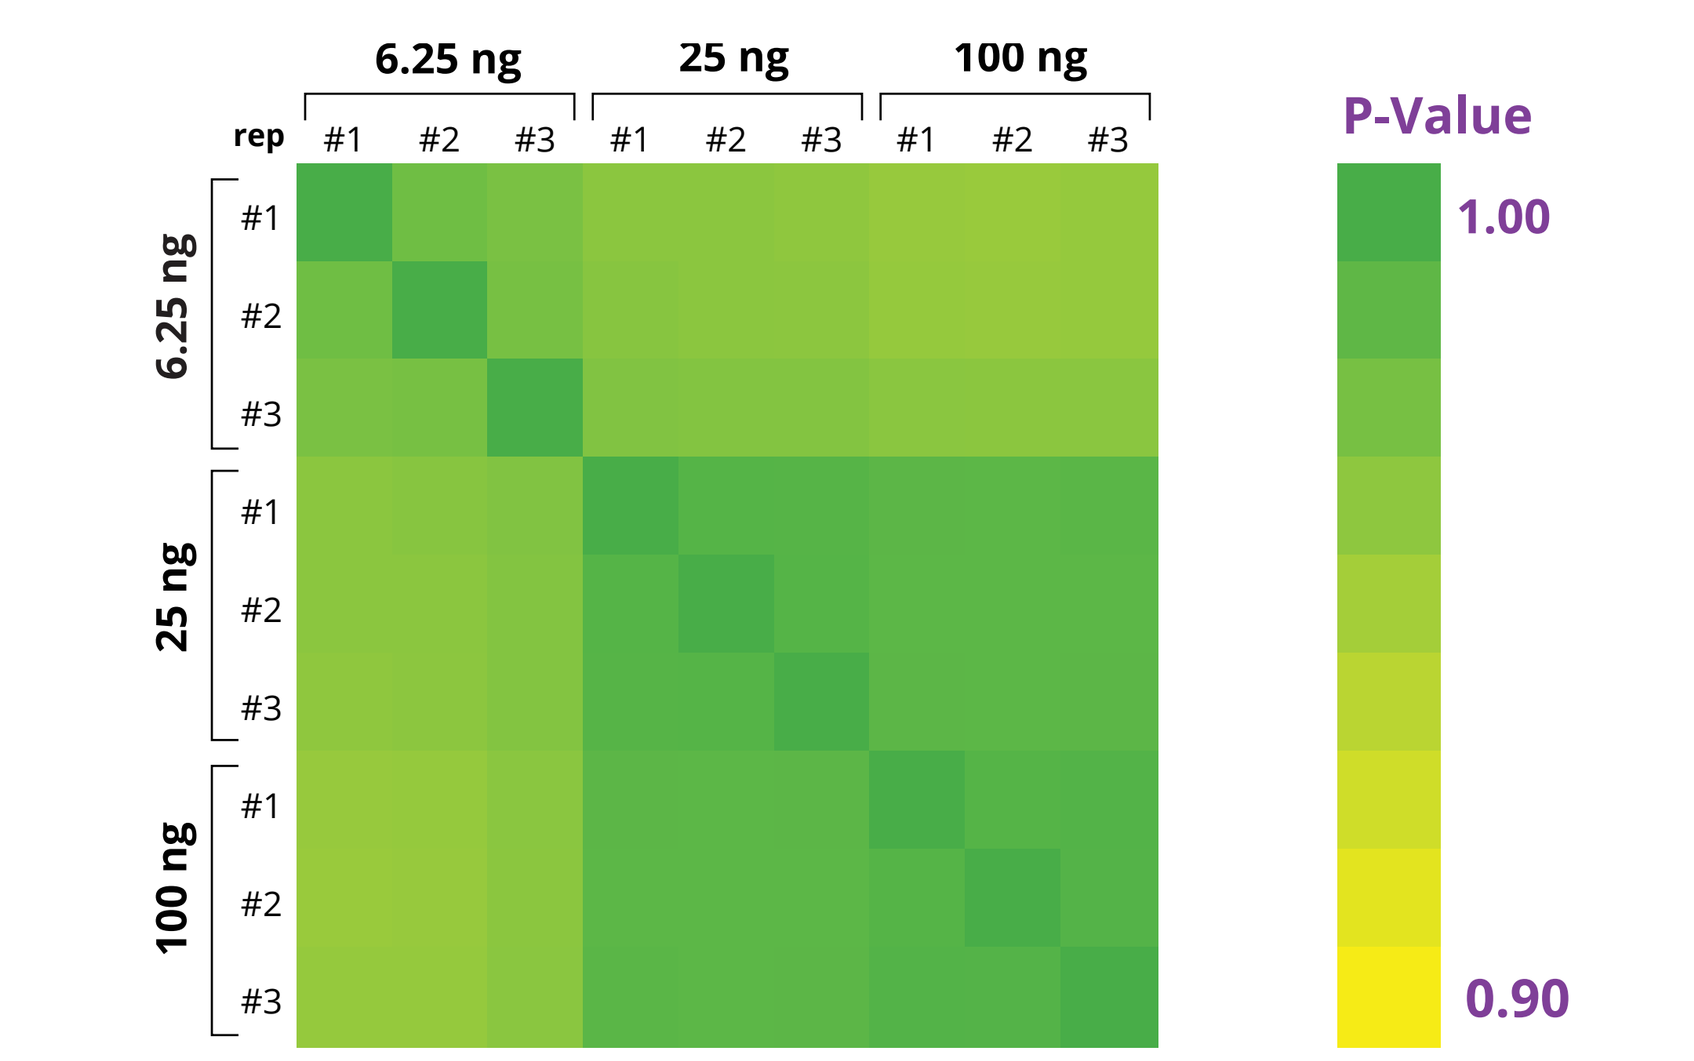
6.25 ng
25 ng
100 ng
rep
P-Value
1.00
0.90

Correlation Matrix of expression profiles at differing amounts of Human Universal RNA (6.25, 25, and 100 ng) in triplicates.

Reproducibility of CancerNet Expression Profiles

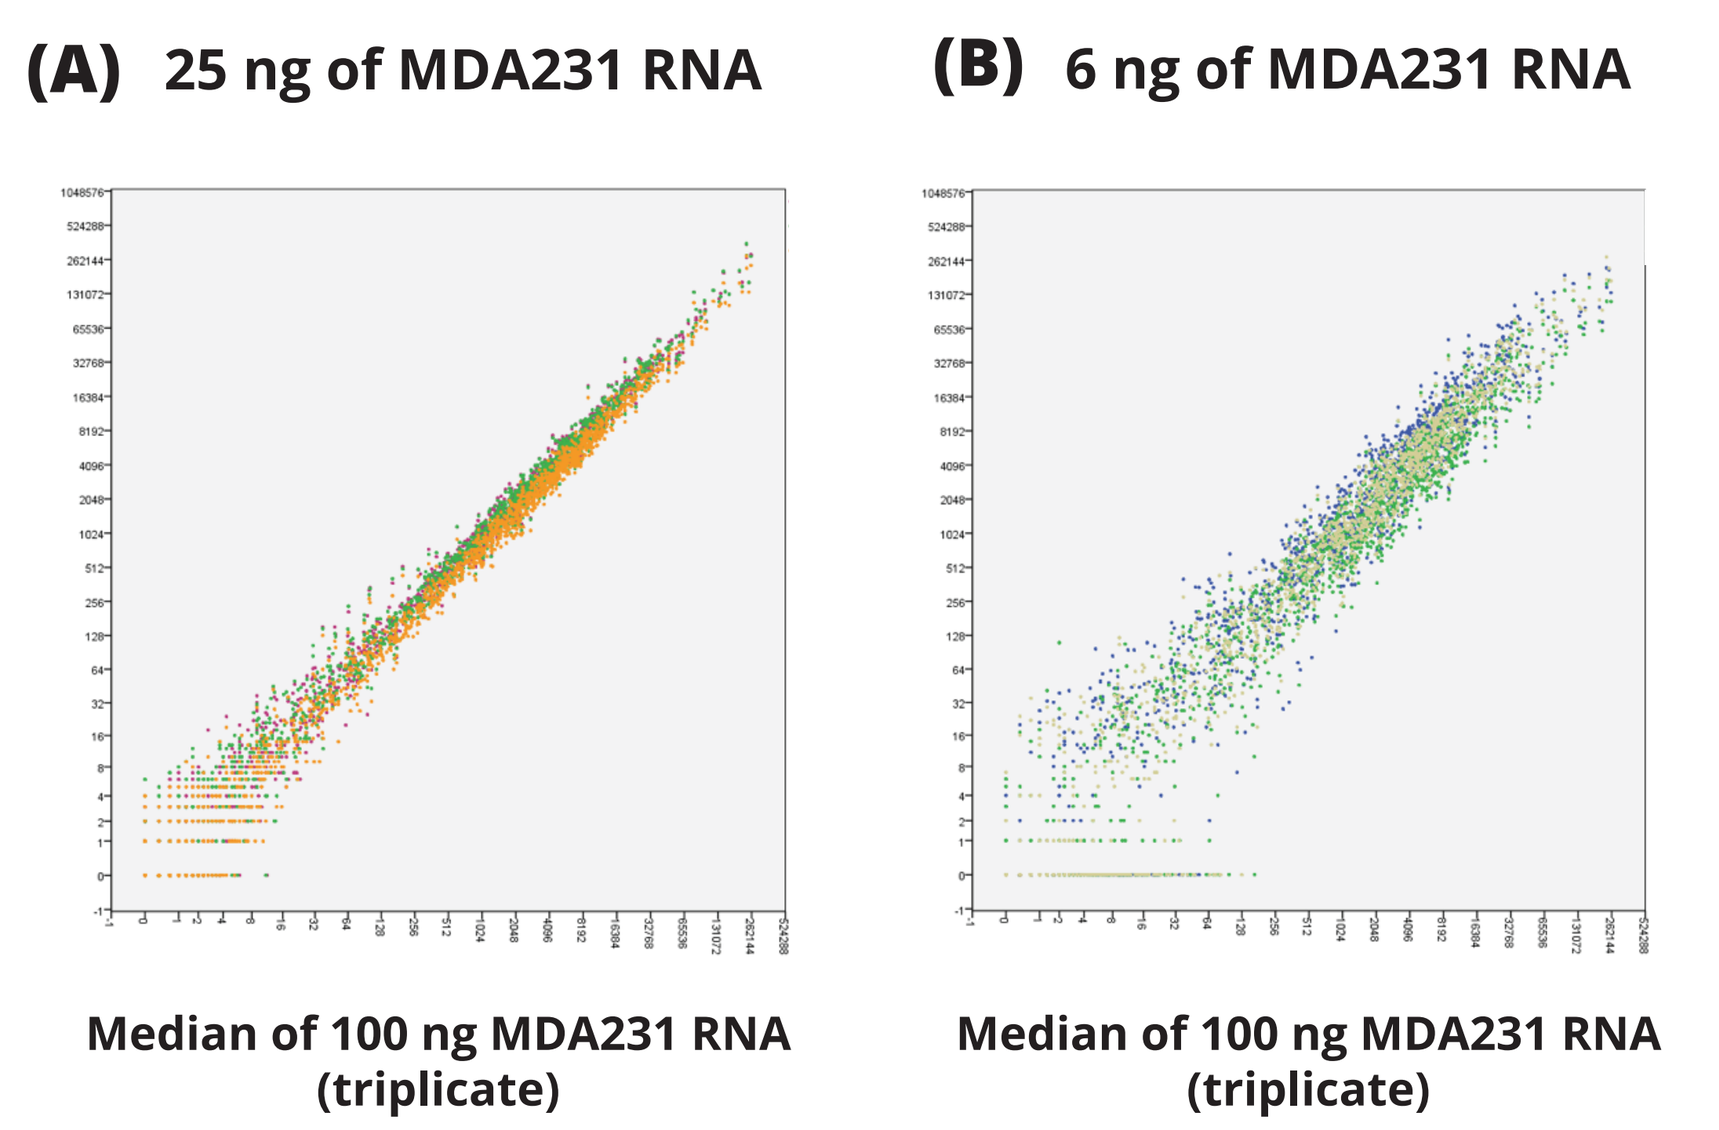
(A) 25 ng of MDA231 RNA
(B) 6 ng of MDA231 RNA
Median of 100 ng MDA231 RNA (triplicate)
Median of 100 ng MDA231 RNA (triplicate)

Scatterplot of CancerNet expression profiles of (A) 25 ng and (B) 6 ng vs. median of 100 ng of MDA231 RNA in triplicate.

Sensitivity and Accuracy of Detection of Immune Cells in Tumor Samples

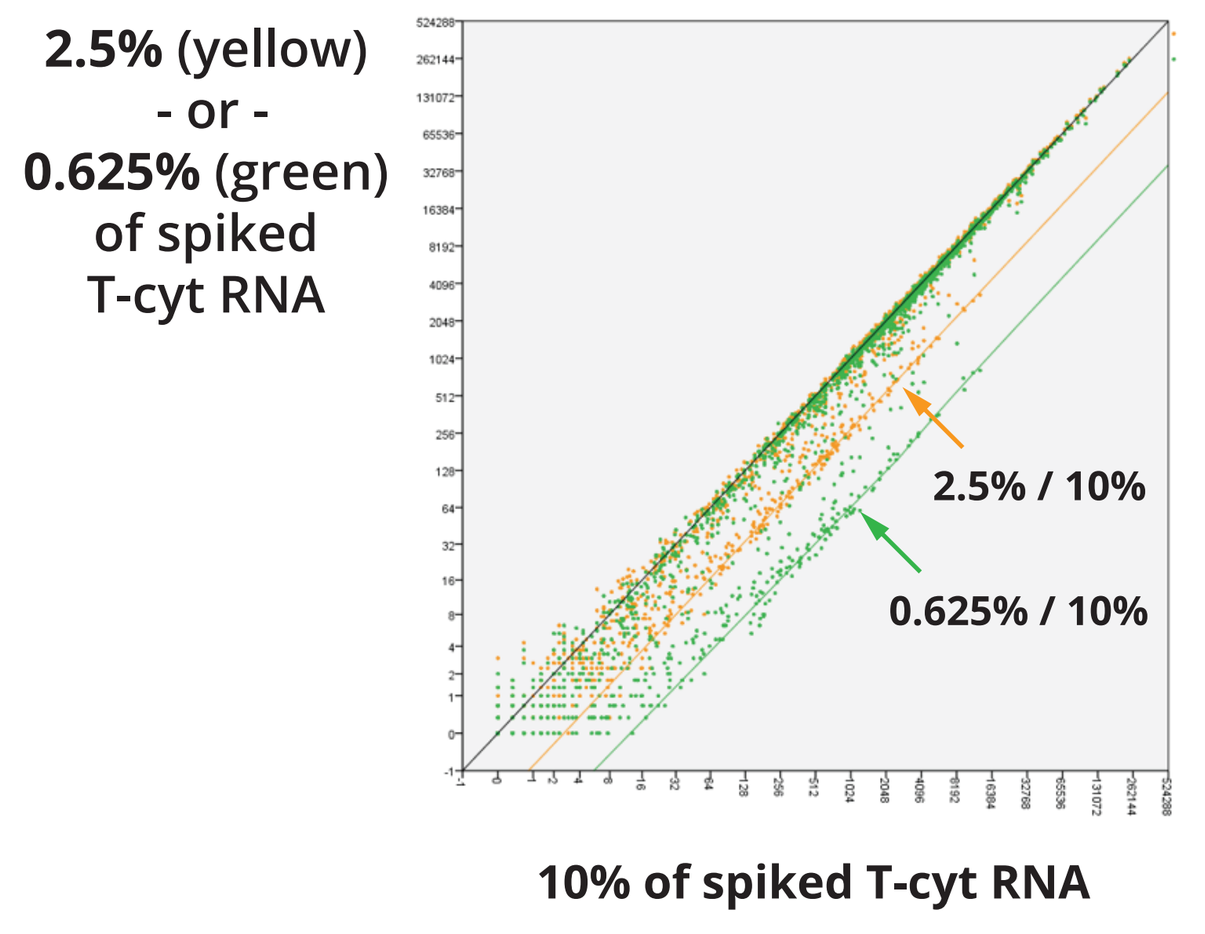
2.5% (yellow)
- or -
0.625% (green)
of spiked
T-cyt RNA
2.5% / 10%
0.625% / 10%
10% of spiked T-cyt RNA

Scatterplot analysis of CancerNet expression profiles generated using a model system of T-cyt RNA spiked at 0.625%, 2.5%, and 10% in 100 ng MDA231 RNA.

Driver-Map™ Portfolio

Assay	Composition	Discovery / Profiling
GeneNet™ 18K *(coming soon)*	General cellular mechanisms Metabolome Housekeeping genes	Generic gene discovery
CancerNet™ 8K	Hallmarks of Cancer Pathway networks Extracellular proteins Drug metabolism	Signaling network mechanisms Cancer biomarkers Cancer Hallmarks Drug resistance mechanisms
Tumor Microenvironment TME 2.5K	Cell-type lineage signatures Immunotherapy & FDA drug targets Immunity mechanisms Immunotherapy biomarkers	Cell composition Immune biomarkers
Cancer Core 540	Pan-cancer driver genes	Mutations in both DNA & RNA

Conclusions

We present the novel CancerNet 8K assay panel, a quantitative, multiplexed, high throughput targeted RNA-Seq approach that leverages the power of multiplex PCR technologies and NGS and allows you to obtain the transcriptome profile of the tumor microenvironment.

- **The CancerNet Panel** includes 8,000 genes, 2,500 of which are immunity-related. In order to develop cell-type specific gene signatures, we developed a non-probabilistic binary linear classifier algorithm to infer the level of infiltrating immune cells in tumor tissues. Our panel can distinguish hematopoietic cell phenotypes from bulk tissues and tumor cells and offers a unique approach to identify tumor-infiltrating cells.
- In this study, we present the design and algorithms of our panel that can accurately resolve relative fractions of diverse immune cell types from complex tissues.

Intended Applications:

- The platform is applicable for novel predictive and prognostic biomarker discovery.
- Comprehensive profiling of tumor-associated immune cell composition will provide important insights into cancer immunoediting mechanisms.
- Has the potential to provide a new molecular stratification approach applicable to cancer immunotherapy.

www.cellecta.com info@cellecta.com Discovery is yours™